A phase I, pharmacokinetic (PK) and preliminary efficacy assessment of ALD518, a humanized anti-IL-6 antibody, in patients with advanced cancer.

S Clarke[1], C. Gebbie[1], C Sweeney[2], N Olszewksi[2], J Smith[3]

[1]Sydney Cancer Centre, Concord Hospital, Sydney, Australia; [2]University of Adelaide, Royal Adelaide Hospital, Adelaide, Australia; [3]Alder Biopharmaceuticals Inc, Bothell, WA, USA

Background

ALD518 is an aglycosylated, humanized, IgG1 antibody that blocks interleukin-6 (IL-6) activity. ALD518 is being developed for the treatment of cancer-related fatigue and cancer cachexia.

Objectives

The primary objective of this study was to determine the safety and tolerability of a single intravenous infusion of ALD518 in patients with advanced cancer.

The secondary objectives were to determine the pharmacokinetics, pharmacodynamics and preliminary efficacy of single infusions of ALD518 in patients with advanced cancer.

Methods

This study was performed in compliance with ICH guidelines for clinical trials and was approved by an independent ethics committee prior to enrollment.

Nine patients with confirmation of a diagnosis of any progressive cancer incurable by other treatments with an ECOG performance status of 0-2, a plasma C-reactive protein concentration ≥ 10mg/L, and at least 18 years of age were enrolled.

Patients received a single intravenous infusion of ALD518 over 1 hour. Three different doses (80mg, 160mg, 320mg) of ALD518 were administered to three cohorts of three patients. The doses were administered in ascending order from 80mg to 320mg with 4 weeks between cohorts. Each cohort was assessed for dose limiting toxicity by an independent Data Monitoring Committee prior to proceeding to the next dose.

Safety data included adverse event monitoring, hematology, clinical chemistry, urinalysis, vital signs, and 12-lead ECGs. Blood samples were taken for pharmcokinetic and pharmacodynamic analysis. The pharmacodynamic biomarker was serum C-reactive protein concentration. Efficacy data included the fatigue subscale of the FACIT-F questionnaire and hand grip strength. Hand grip strength was measured using a hand dynamometer. Patients were followed up from screening to 12 weeks post-dose.

Statistical analysis (ITT) using a non-parametric method (Wilcoxon rank sum test) with last observation carried forward was performed on the hemoglobin, platelet count, neutrophil count, plasma albumin concentration, FACIT-F questionnaire, hand grip strength, and C-reactive protein. Every patient with more than one post-dose value (8 out of 9) was included in the analysis.

Baseline Patient Demographics

Mean (SD)	80mg ALD518 (n=3)	160 mg ALD518 (n=3)	320mg ALD518 (n=3)
Age (years)	74.3 (8.7)	67.5 (17.1)	73.1 (9.8)
Weight (kg)	73.2 (17.9)	71.6 (14.0)	80.8 (8.8)
Height (cm)	164.0 (7.2)	163.7 (15.7)	170.0 (6.6)

Baseline Patient Malignancy History

	80mg ALD518 (n=3)	160 mg ALD518 (n=3)	320mg ALD518 (n=3)
Primary:			
Colorectal	2	1	1
Cholangiocarcinoma	1	1	-
Non-small cell lung	-	1	-
Mesothelioma	-	-	2
ECOG status:			
0	-	-	-
1	2	2	1
2	1	1	2
Metastatic disease:			
Yes	3	3	3
No	-	-	-
Prior chemotherapy:			
Yes	3	1	3
No	0	2	-
Prior radiotherapy:			
Yes	1	-	2
No	2	3	1

Results - Safety

All patients were evaluable for DLT assessment, although 4 patients failed to complete all study procedures due to progressive malignancy. 1 patient on the 80mg dose withdrew after the week 1 visit, 1 patient on the 160mg dose withdrew after the week 8 visit, and two patients withdrew on the 320mg dose after the week 8 and week 10 visit, respectively.

There were 4 serious adverse events during the study: 3 disease progressions and 1 sepsis secondary to a blocked biliary stent. The most common adverse events, occurring in at least 25% of subjects were nausea, vomiting, abdominal pain, diarrhea, gastro esophageal reflux disease, and disease progression. There were no infusion reactions noted in any patient.

Results - Safety

For the laboratory safety parameters, efficacy, and CRP discussed below 8 of the 9 patients in the study were evaluable for the ITT analysis. All p values in the graphs are versus baseline (pre-dose) for that time-point.

Hemoglobin increased in 7 of the 8 patients and this increase was maintained throughout the 12 weeks of the study (figure 1).

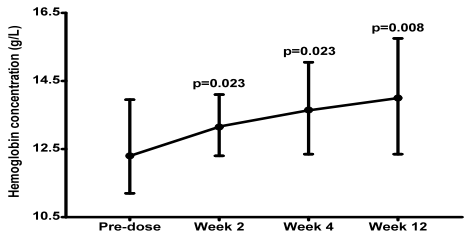
Hemoglobin concentration (g/L)
16.5
14.5
12.5
10.5
p=0.023
p=0.023
p=0.008
Pre-dose
Week 2
Week 4
Week 12

Figure 1: Median and interquartile range for hemoglobin concentration (n=8).

Plasma albumin concentration increased in all 8 patients and this increase was maintained throughout the 12 weeks of the study (figure 2).

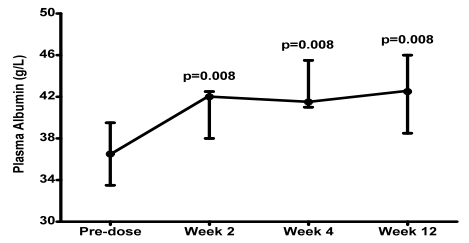
Plasma Albumin (g/L)
50
46
42
38
34
30
p=0.008
p=0.008
p=0.008
Pre-dose
Week 2
Week 4
Week 12

Figure 2: Median and interquartile range for plasma albumin concentration (n=8).

Platelet counts decreased in all 8 patients and stabilized out to week 12 (figure 3).

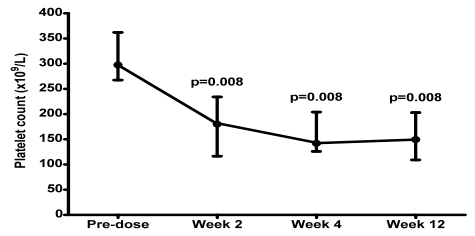
Platelet count (x10^9/L)
400
350
300
250
200
150
100
50
0
p=0.008
p=0.008
p=0.008
Pre-dose
Week 2
Week 4
Week 12

Figure 3: Median and interquartile range for platelet count (n=8).

Neutrophil counts decreased in all 8 patients and stabilized out to week 12 (figure 4).

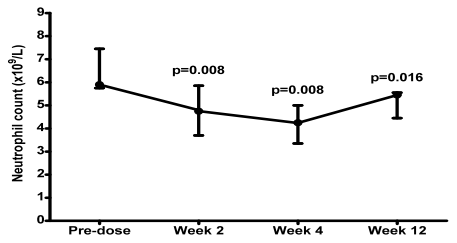
Neutrophil count (x10^9/L)
9
8
7
6
5
4
3
2
1
0
p=0.008
p=0.008
p=0.016
Pre-dose
Week 2
Week 4
Week 12

Figure 4: Median and interquartile range for neutrophil count (n=8).

Results - Efficacy

There was a reduction in fatigue in 7 out of the eight patients and this reduction was maintained out to week 12 as judged by the FACIT-F questionnaire (figure 5).

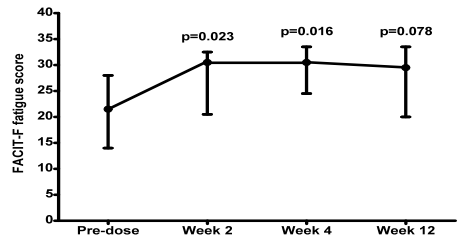
FACIT-F fatigue score
40
35
30
25
20
15
10
5
0
p=0.023
p=0.016
p=0.078
Pre-dose
Week 2
Week 4
Week 12

Figure 5: Median and interquartile range for FACIT-F fatigue score (n=8).

There was an improvement in hand grip strength in 5 out of 8 of the patients (figure 6).

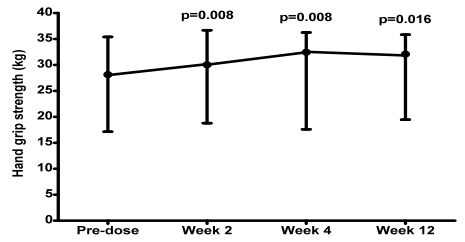
Hand grip strength (kg)
40
35
30
25
20
15
10
5
0
p=0.008
p=0.008
p=0.016
Pre-dose
Week 2
Week 4
Week 12

Figure 6: Median and interquartile range for hand grip strength (n=8).

Results – PK/PD

The PK of ALD518 were linear and dose proportional (in terms of AUC_{0-t} and C_{max}) for all the doses. The median elimination half-life of ALD518 was 25 days (inter quartile range: 20 – 32 days).

Serum CRP concentrations decreased and remained very low throughout the 12 week follow-up period in all 8 patients (figure 7).

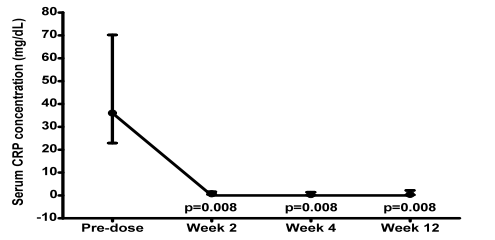
Serum CRP concentration (mg/dL)
80
70
60
50
40
30
20
10
0
-10
p=0.008
p=0.008
p=0.008
Pre-dose
Week 2
Week 4
Week 12

Figure 7: Median and interquartile range for serum C-reactive protein concentration (n=8).

Conclusions

ALD518 given to patients with advanced cancer was safe and well tolerated. ALD518 reversed fatigue, increased hemoglobin and albumin, and there was a trend to increased hand grip strength. There was a decrease in platelet and neutrophil count that remained stable throughout the study. The median elimination half life of ALD518 was 25 days. There was complete suppression of serum CRP at all doses at all time-points post-dose. The 80mg, 160mg, and 320mg doses have been taken forward into a multiple dose 6 month phase II study in NSCLC patients with cachexia.